Widespread CNS Tau Knockdown for the Potential Treatment of Alzheimer's Disease and Other Tauopathies

Bryan Zeitler[1], Kimberly Marlen[1], David Ojala[1], Annemarie Ledeboer[1], Qi Yu[1], Yonghua Pan[1], Marina Falaleeva[1], Giulia Cisbani[2], Finn Peters[2], Chiara Melis[2], Sarah Mueller[1], Matthew Tiffany[1], Kenneth Kennard[1], Patrick Dunn[1], Hoang-Oanh Nguyen[1], Hung Tran[1], Kathleen Meyer[1], Amy M Pooler[1]

[1] Sangamo Therapeutics, Inc., Richmond, CA
[2] Evotec SE, Hamburg, Germany

Sangamo THERAPEUTICS™

Poster #1126

Introduction

- The neuronal accumulation of misfolded, neurotoxic species of Microtubule Associated Protein Tau (*MAPT*) closely tracks with neurodegeneration, disease progression, and clinical symptoms in Alzheimer's disease (AD), Progressive Supranuclear Palsy (PSP) and over a dozen other tauopathy disorders.
- Reducing *MAPT* expression is well-tolerated and efficacious in tauopathy animal models. We are developing an IV-administered, epigenetic regulation approach to reduce tau expression using an engineered zinc finger repressor (ZFR).
- Here, we report the development of a lead ZFR that targets the human *MAPT* locus with high potency and specificity across several in vitro and in vivo models.
- We also show widespread central nervous system (CNS) ZFR expression and *MAPT* knockdown in adult nonhuman primates (NHPs) following a single IV administration of STAC-BBB, an AAV capsid that we have engineered to cross the blood-brain barrier (BBB).

A ZFR genomic medicine for the potential treatment of AD and other tauopathies

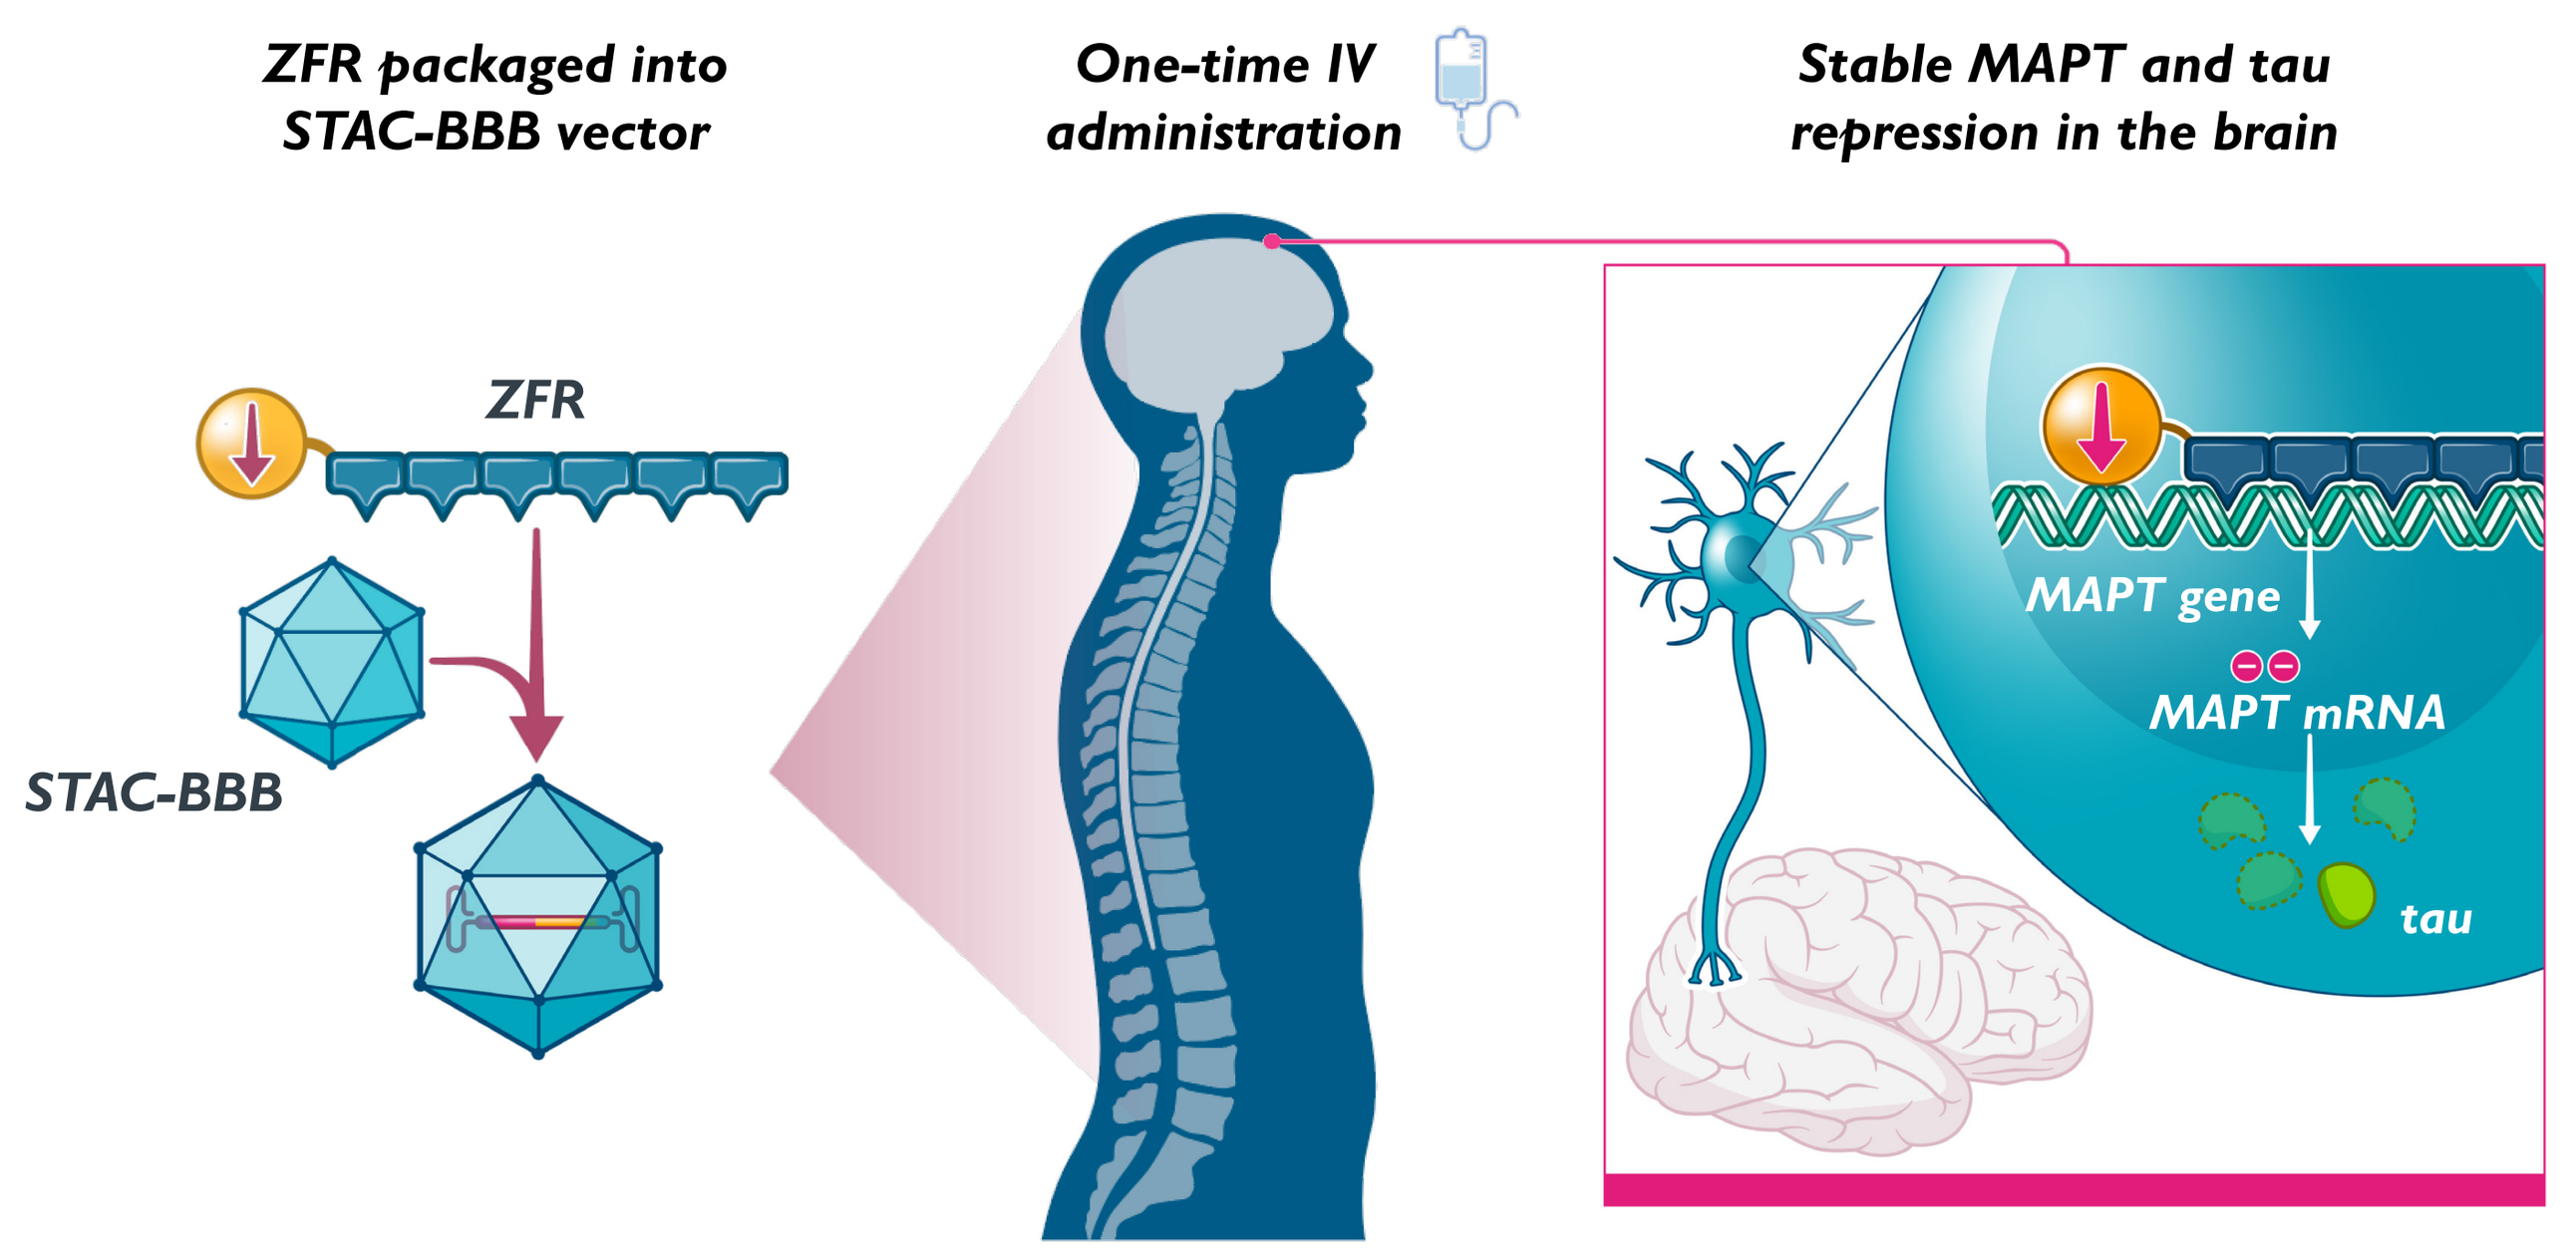
ZFR packaged into STAC-BBB vector
One-time IV administration
Stable MAPT and tau repression in the brain
ZFR
STAC-BBB
MAPT gene
MAPT mRNA
tau

Figure 1. AAV ZFR for tauopathies

A ZFR targeting the tau gene is packaged into STAC-BBB, which crosses the BBB with a one-time IV administration. The ZFR stably represses *MAPT* transcription in neurons, resulting in specific reduction of tau protein. Lowering neuronal tau allows for clearance of all aberrant tau protein forms, improved neuron function and survival, and reduced susceptibility to tau mediated toxicity.

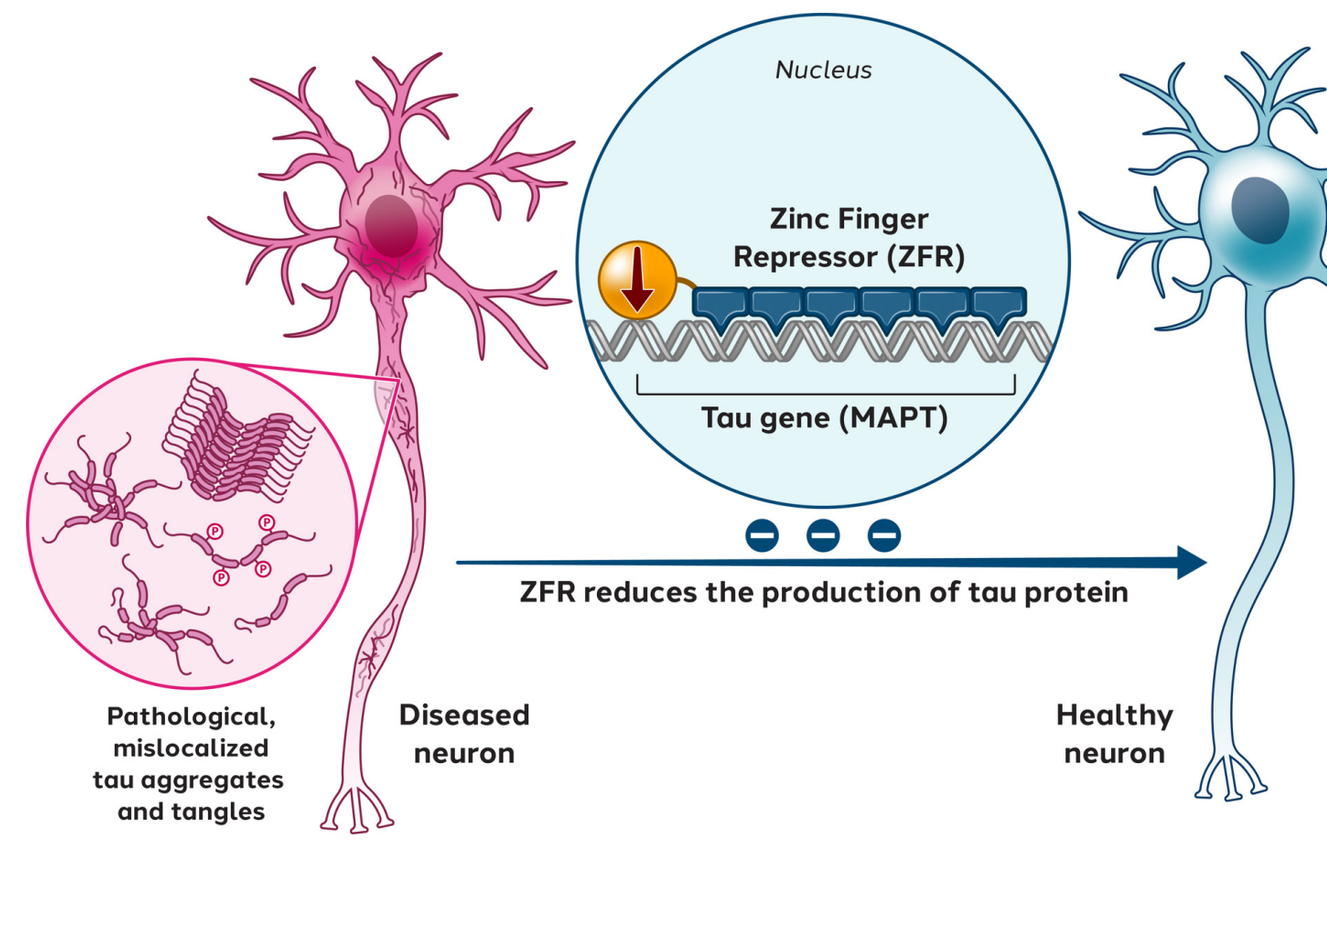
Nucleus
Zinc Finger Repressor (ZFR)
Tau gene (MAPT)
ZFR reduces the production of tau protein
Pathological, mislocalized tau aggregates and tangles
Diseased neuron
Healthy neuron

Conclusions and next steps

- We characterized a candidate ZFR that represses human *MAPT* by >95% with no off targets.
- Potent ZFR-mediated *MAPT* repression was demonstrated at the bulk and single-cell level in the mouse and NHP CNS. ZFR treatment reduced total tau and p-tau in htau mice.
- ZFR treatment was well tolerated in all models, doses, and time points tested.
- This work supports the initiation of an IND-enabling GLP toxicology study for a STAC-BBB delivered ZFR for the potential treatment of tauopathies, including AD and PSP.

Highly potent and specific ZFRs targeting *MAPT*

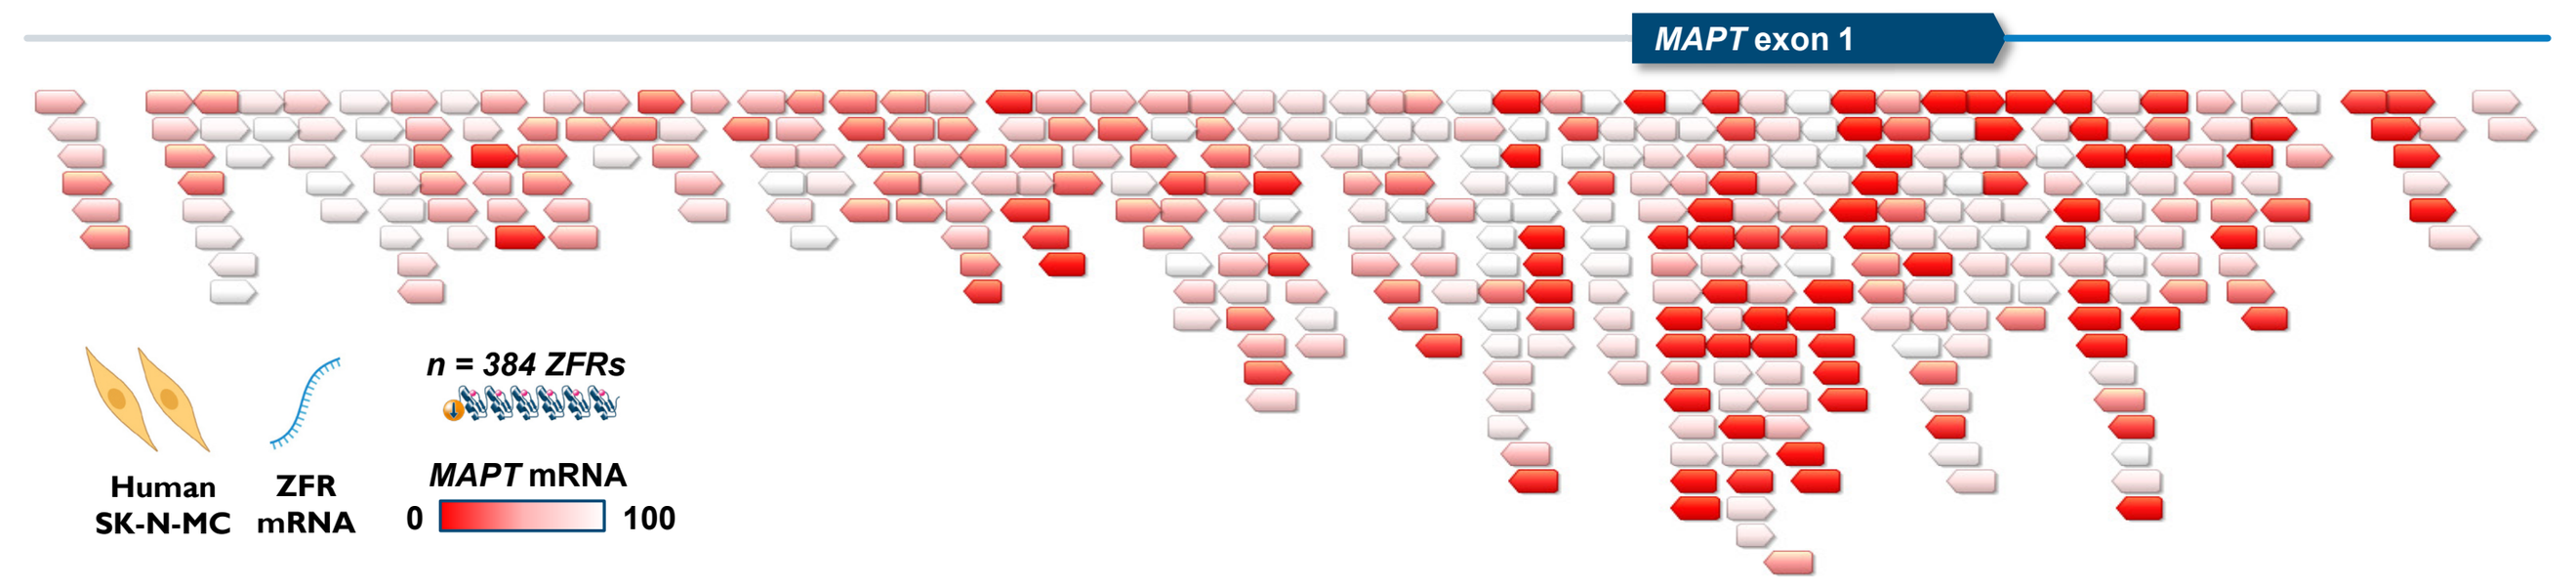
MAPT exon 1
n = 384 ZFRs
Human SK-N-MC
ZFR mRNA
MAPT mRNA
0
100

Figure 2. Initial screen for ZFRs targeting human *MAPT*

384 ZFRs were designed to target human *MAPT* and screened by transient transfection in SK-N-MC cells. ~29% of ZFRs achieved at least 50% repression of the *MAPT* transcript.

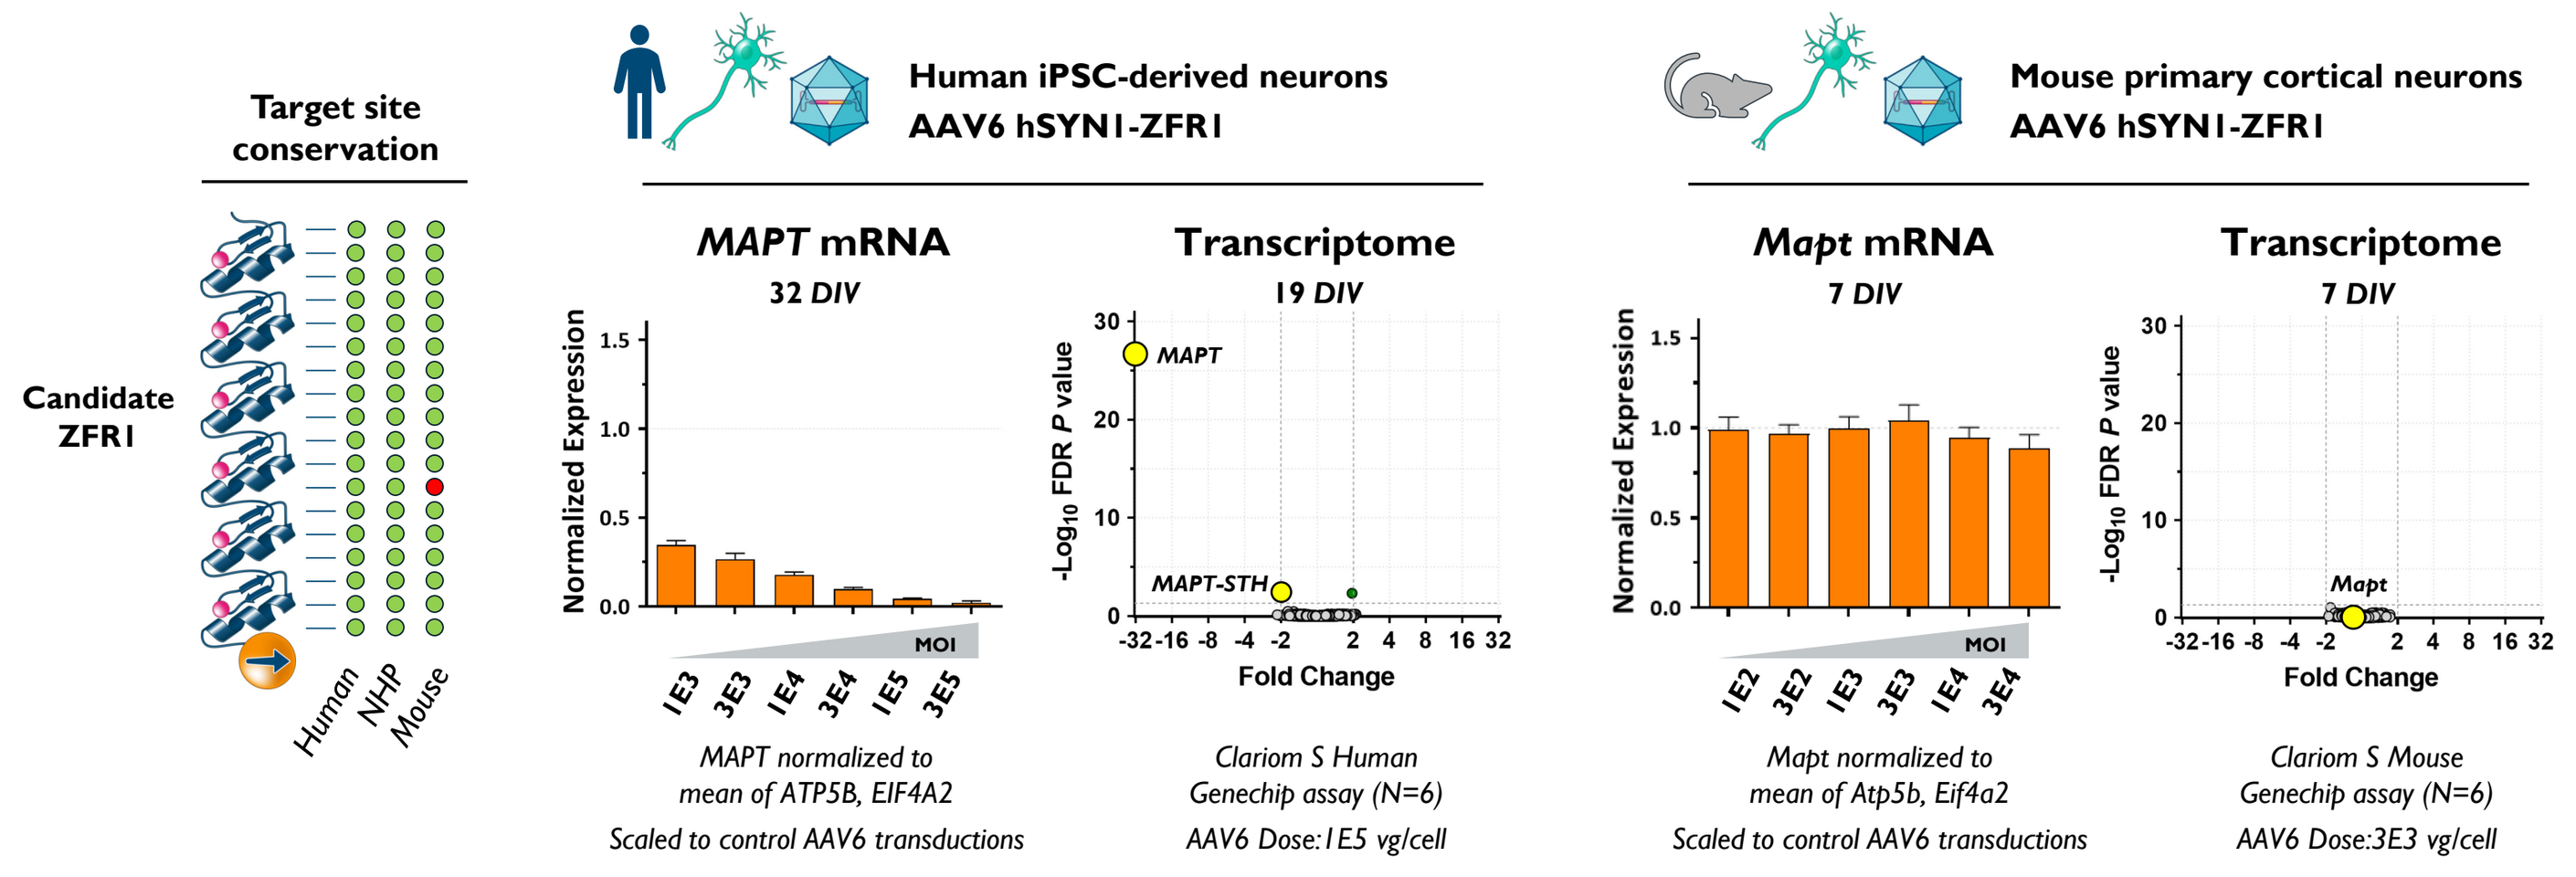
Target site conservation
Candidate ZFR1
Human
NHP
Mouse
Human iPSC-derived neurons
AAV6 hSYN1-ZFR1
MAPT mRNA
32 DIV
Normalized Expression
MOI
MAPT normalized to mean of ATP5B, EIF4A2
Scaled to control AAV6 transductions
Transcriptome
19 DIV
MAPT
MAPT-STH
Fold Change
Clariom S Human Genechip assay (N=6)
AAV6 Dose:1E5 vg/cell
Mouse primary cortical neurons
AAV6 hSYN1-ZFR1
Mapt mRNA
7 DIV
Mapt normalized to mean of Atp5b, Eif4a2
Scaled to control AAV6 transductions
Transcriptome
7 DIV
Mapt
Clariom S Mouse Genechip assay (N=6)
AAV6 Dose:3E3 vg/cell

Figure 3. Characterization of a potent, specific ZFR candidate targeting human *MAPT*

ZFR1 reduced *MAPT* in human iPSC-derived neurons by >95% with no off-targets. In mouse primary cortical neurons, ZFR1 was highly specific, but showed no repression of mouse *Mapt* due to a single mismatch in the mouse target site.

ZFR1 stably lowers tau mRNA and protein by >90% in vivo after hippocampal delivery to htau mice

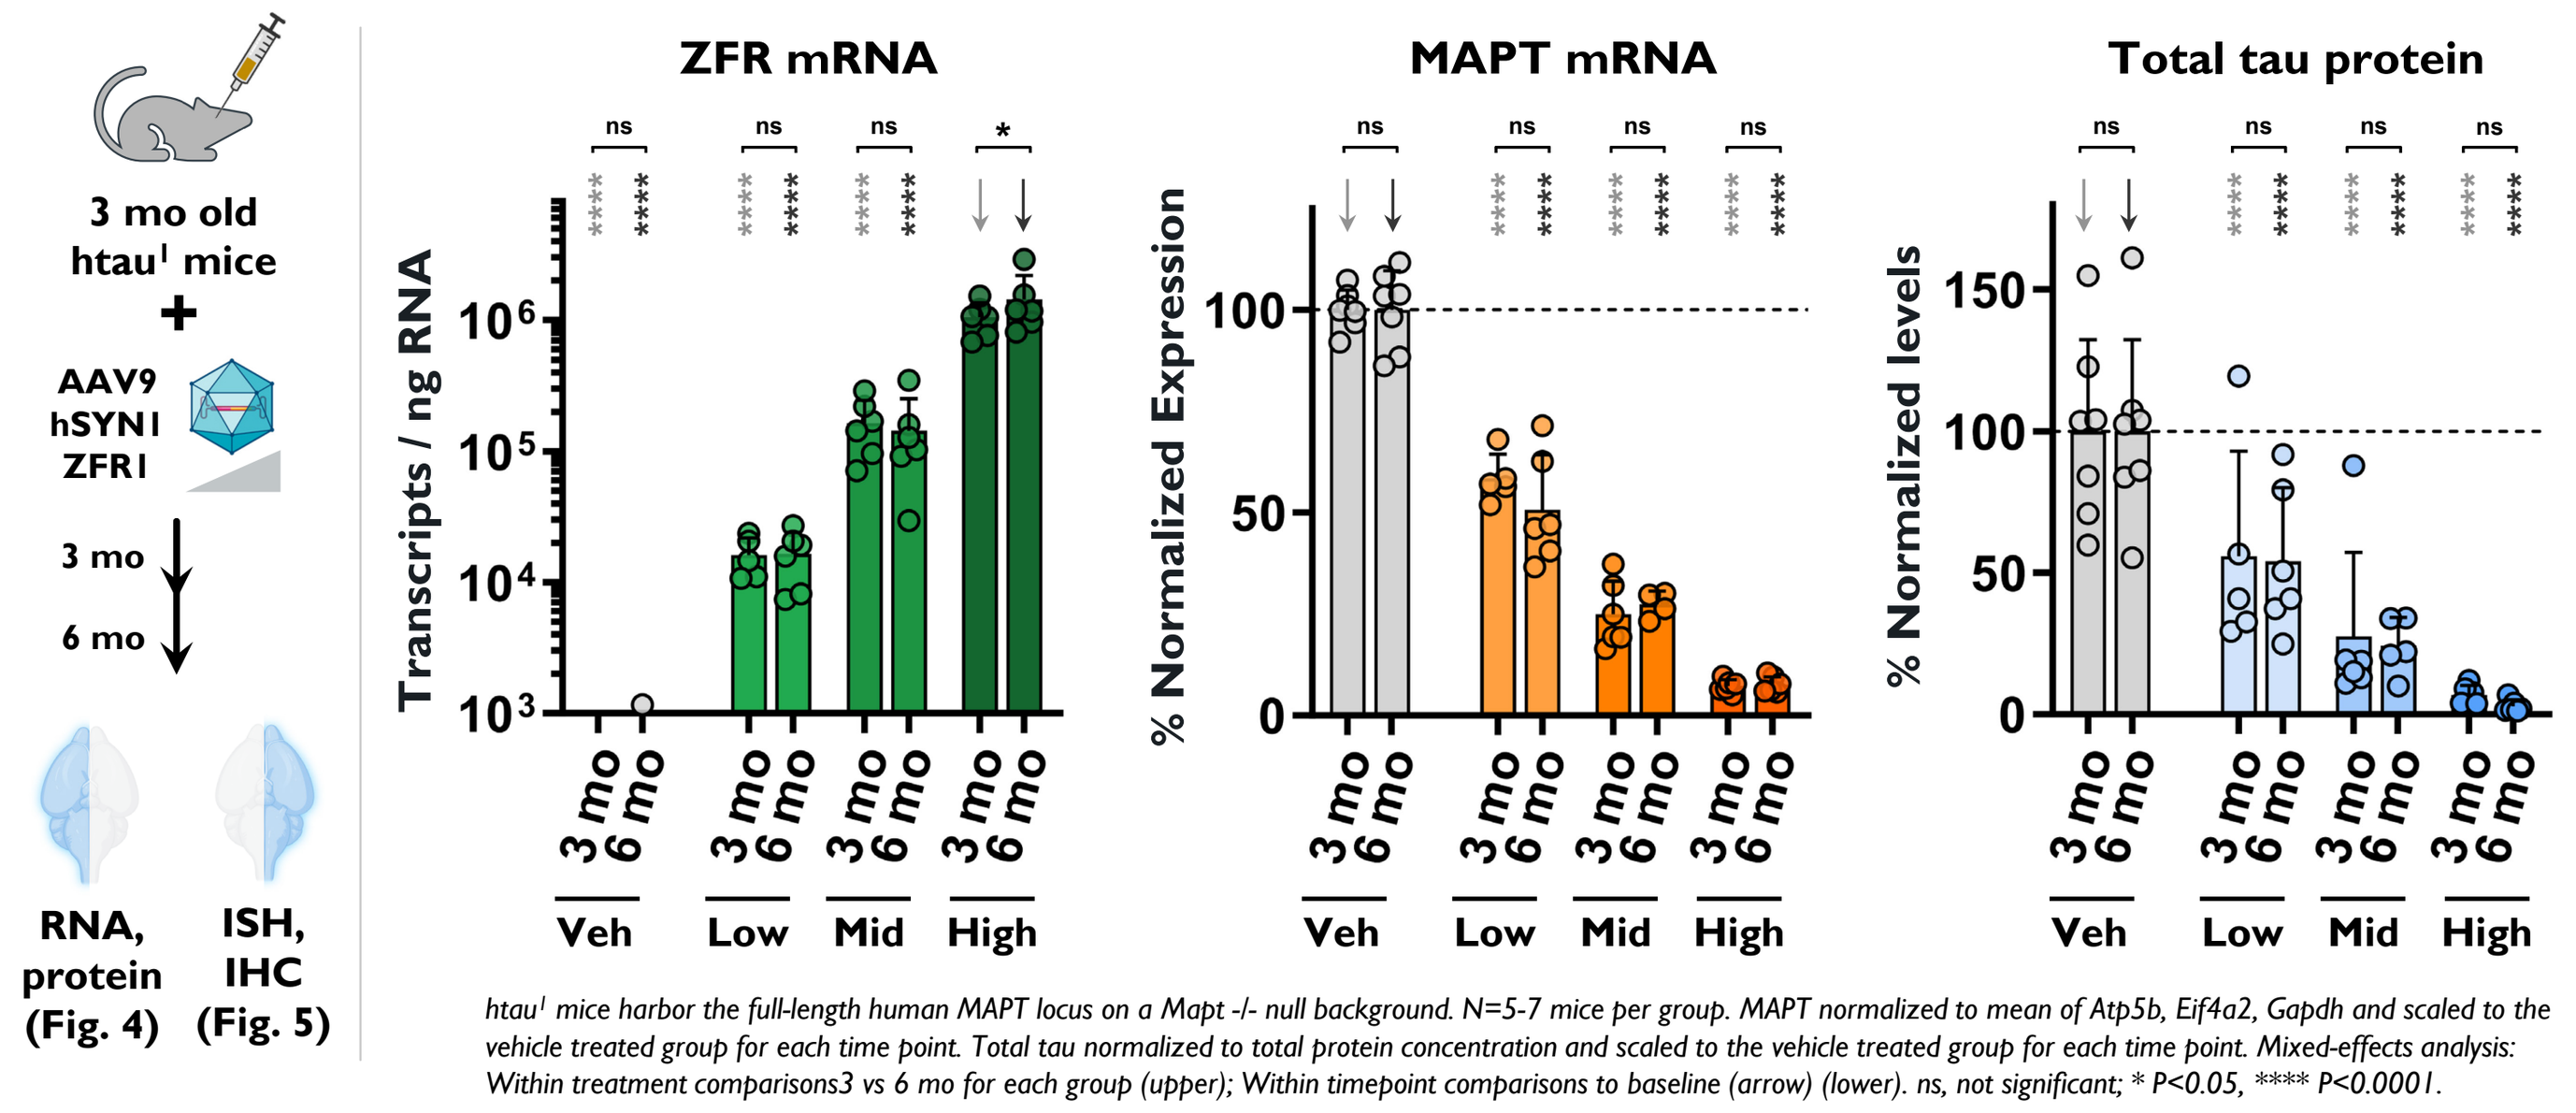
3 mo old htau[1] mice
AAV9 hSYN1 ZFR1
3 mo
6 mo
RNA, protein (Fig. 4)
ISH, IHC (Fig. 5)
ZFR mRNA
Transcripts / ng RNA
MAPT mRNA
% Normalized Expression
Total tau protein
% Normalized levels
Veh
Low
Mid
High
htau[1] mice harbor the full-length human MAPT locus on a Mapt -/- null background. N=5-7 mice per group. MAPT normalized to mean of Atp5b, Eif4a2, Gapdh and scaled to the vehicle treated group for each time point. Total tau normalized to total protein concentration and scaled to the vehicle treated group for each time point. Mixed-effects analysis: Within treatment comparisons3 vs 6 mo for each group (upper); Within timepoint comparisons to baseline (arrow) (lower). ns, not significant; * P<0.05, **** P<0.0001.

Figure 4. Reduction of human *MAPT* in the hippocampus of htau transgenic mice

htau[1] mice received dual, bilateral intrahippocampal injections of AAV9 hSYN1-ZFR1 at 3E8, 3E9, or 3E10 VG per hemisphere. Cohorts of mice were euthanized at 3 mo or 6 mo post injection. A clear dose response for ZFR expression, *MAPT* repression, and total tau protein knockdown was observed, ranging from ~50 to >90% at both the 3 mo and 6 mo timepoints. The treatment was well tolerated and there were no test-article related histopathology findings.

[1] Andorfer C. et al. J Neurochem. 2003;86(3):582-590.

ZFR1 reduces *MAPT* expression at the single-neuron level over a 100-fold dose range in vivo

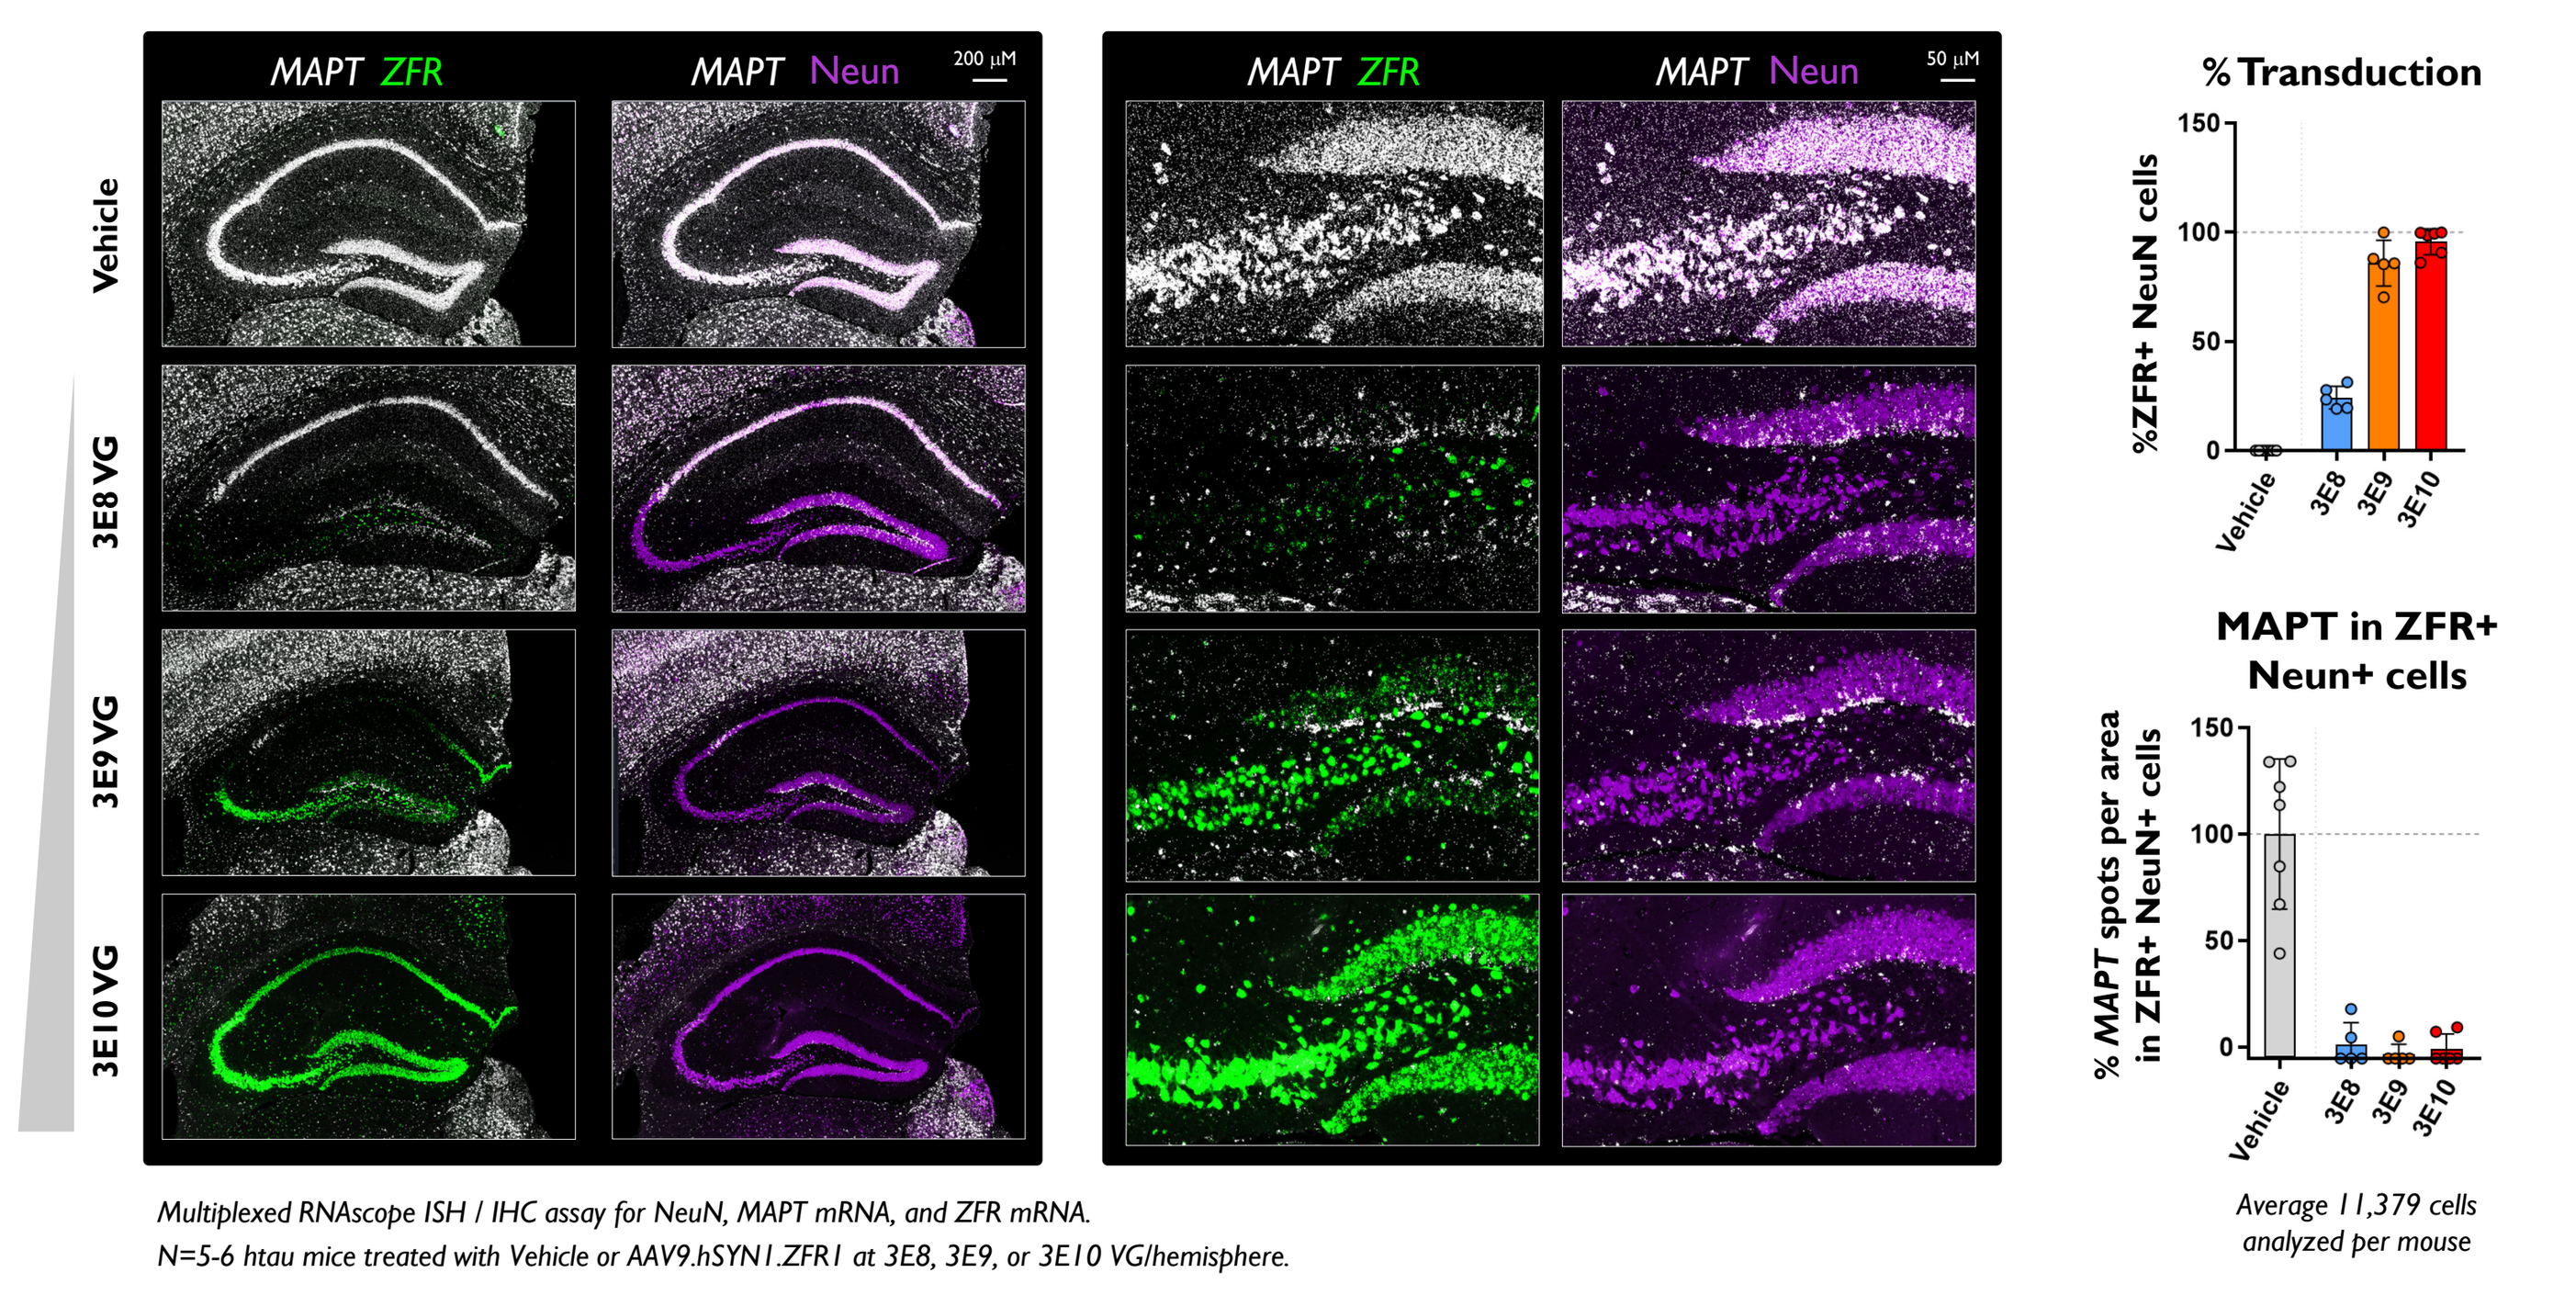
MAPT ZFR
MAPT Neun
Vehicle
3E8 VG
3E9 VG
3E10 VG
% Transduction
%ZFR+ NeuN cells
MAPT in ZFR+ Neun+ cells
% MAPT spots per area in ZFR+ NeuN+ cells
Average 11,379 cells analyzed per mouse
Multiplexed RNAscope ISH / IHC assay for NeuN, MAPT mRNA, and ZFR mRNA.
N=5-6 htau mice treated with Vehicle or AAV9.hSYN1.ZFR1 at 3E8, 3E9, or 3E10 VG/hemisphere.

Figure 5. Single-cell analysis of ZFR+ neurons in the hippocampus of htau mice

A dose response for neuronal transduction and hippocampal coverage was observed across a 100-fold AAV dose range as assessed by multiplexed ISH/IHC (see Fig. 4 for study details). At all doses tested, individual neurons expressing ZFR1 showed >95% human *MAPT* reduction. ZFR1 therefore achieves potent single-neuron *MAPT* knockdown over a wide range of expression levels.

Widespread knockdown of human *MAPT* following IV delivery of ZFR1 to aged htau mice

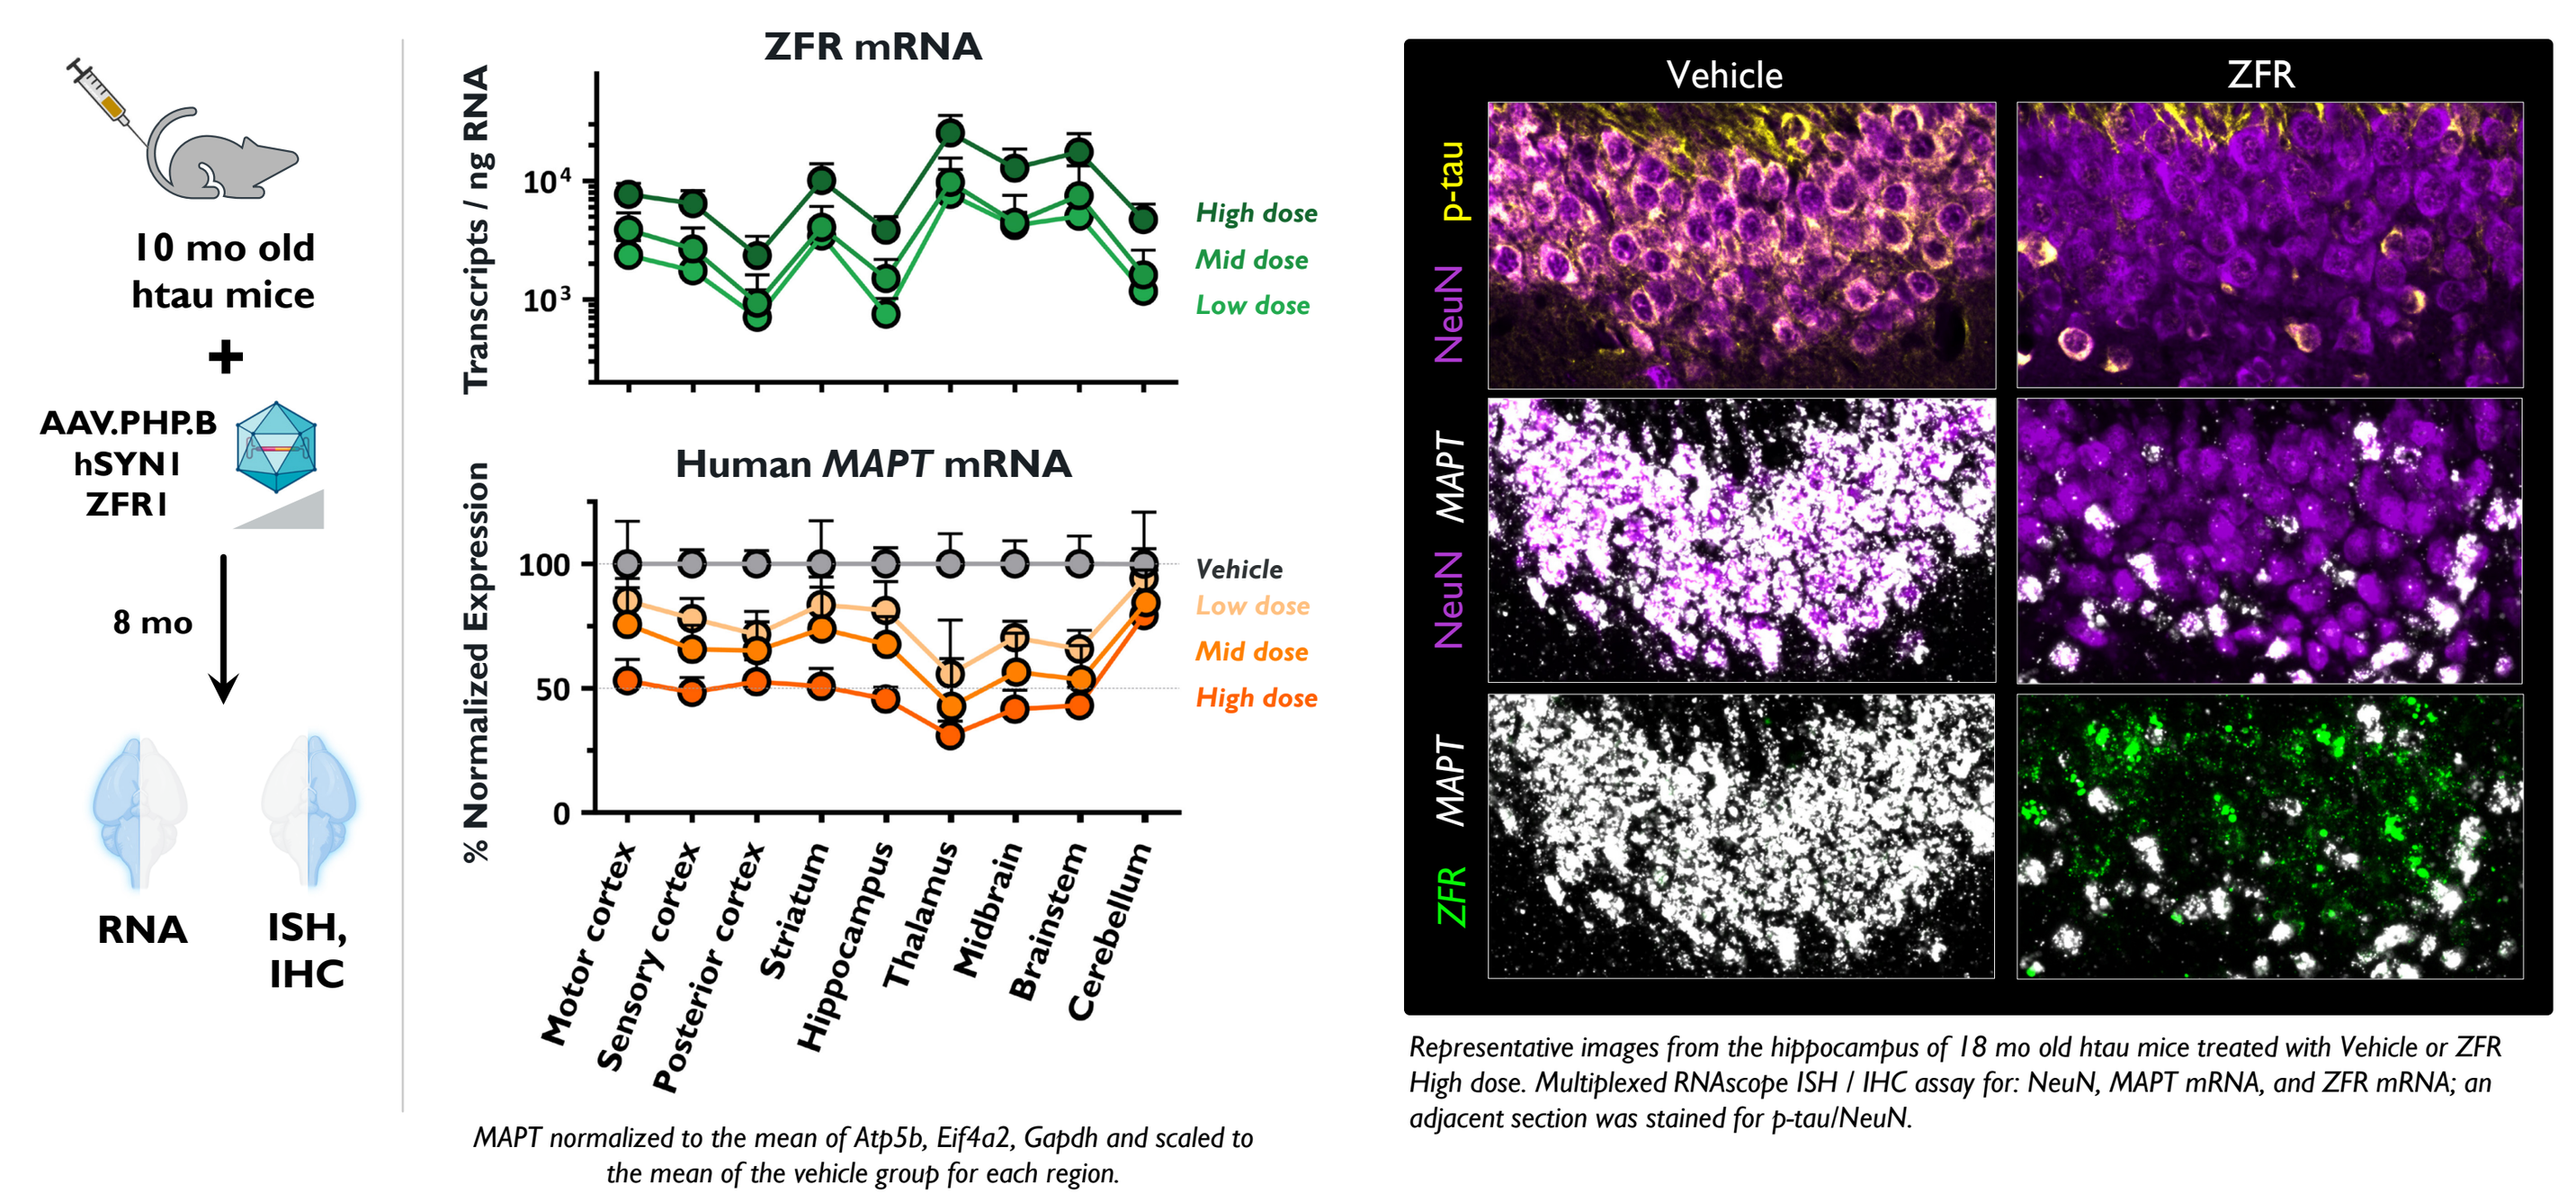
10 mo old htau mice
AAV.PHP.B hSYN1 ZFR1
8 mo
RNA
ISH, IHC
ZFR mRNA
Transcripts / ng RNA
High dose
Mid dose
Low dose
Human MAPT mRNA
% Normalized Expression
Vehicle
Motor cortex
Sensory cortex
Posterior cortex
Striatum
Hippocampus
Thalamus
Midbrain
Brainstem
Cerebellum
MAPT normalized to the mean of Atp5b, Eif4a2, Gapdh and scaled to the mean of the vehicle group for each region.
Vehicle
ZFR
p-tau
NeuN
MAPT
ZFR
Representative images from the hippocampus of 18 mo old htau mice treated with Vehicle or ZFR High dose. Multiplexed RNAscope ISH / IHC assay for: NeuN, MAPT mRNA, and ZFR mRNA; an adjacent section was stained for p-tau/NeuN.

Figure 6. Reduction of human *MAPT* throughout the brain of aged htau mice

10 mo old htau mice received tail vein injections of AAV.PHP.B hSYN1-ZFR1 at 1.2E13, 3.5E13, or 1.2E14 VG/kg. Mice were euthanized 8 mo after injection. A dose response for ZFR expression and *MAPT* repression was observed in all 9 brain regions analyzed, with ~50-70% reduction at the highest dose in most regions. Single-cell analysis using multiplexed ISH/IHC revealed potent *MAPT* repression across the brain, including reduction of hyperphosphorylated p-tau in hippocampal neurons. The treatment was well tolerated and there were no test-article related histopathology findings.

STAC-BBB enables whole CNS tau knockdown in NHPs

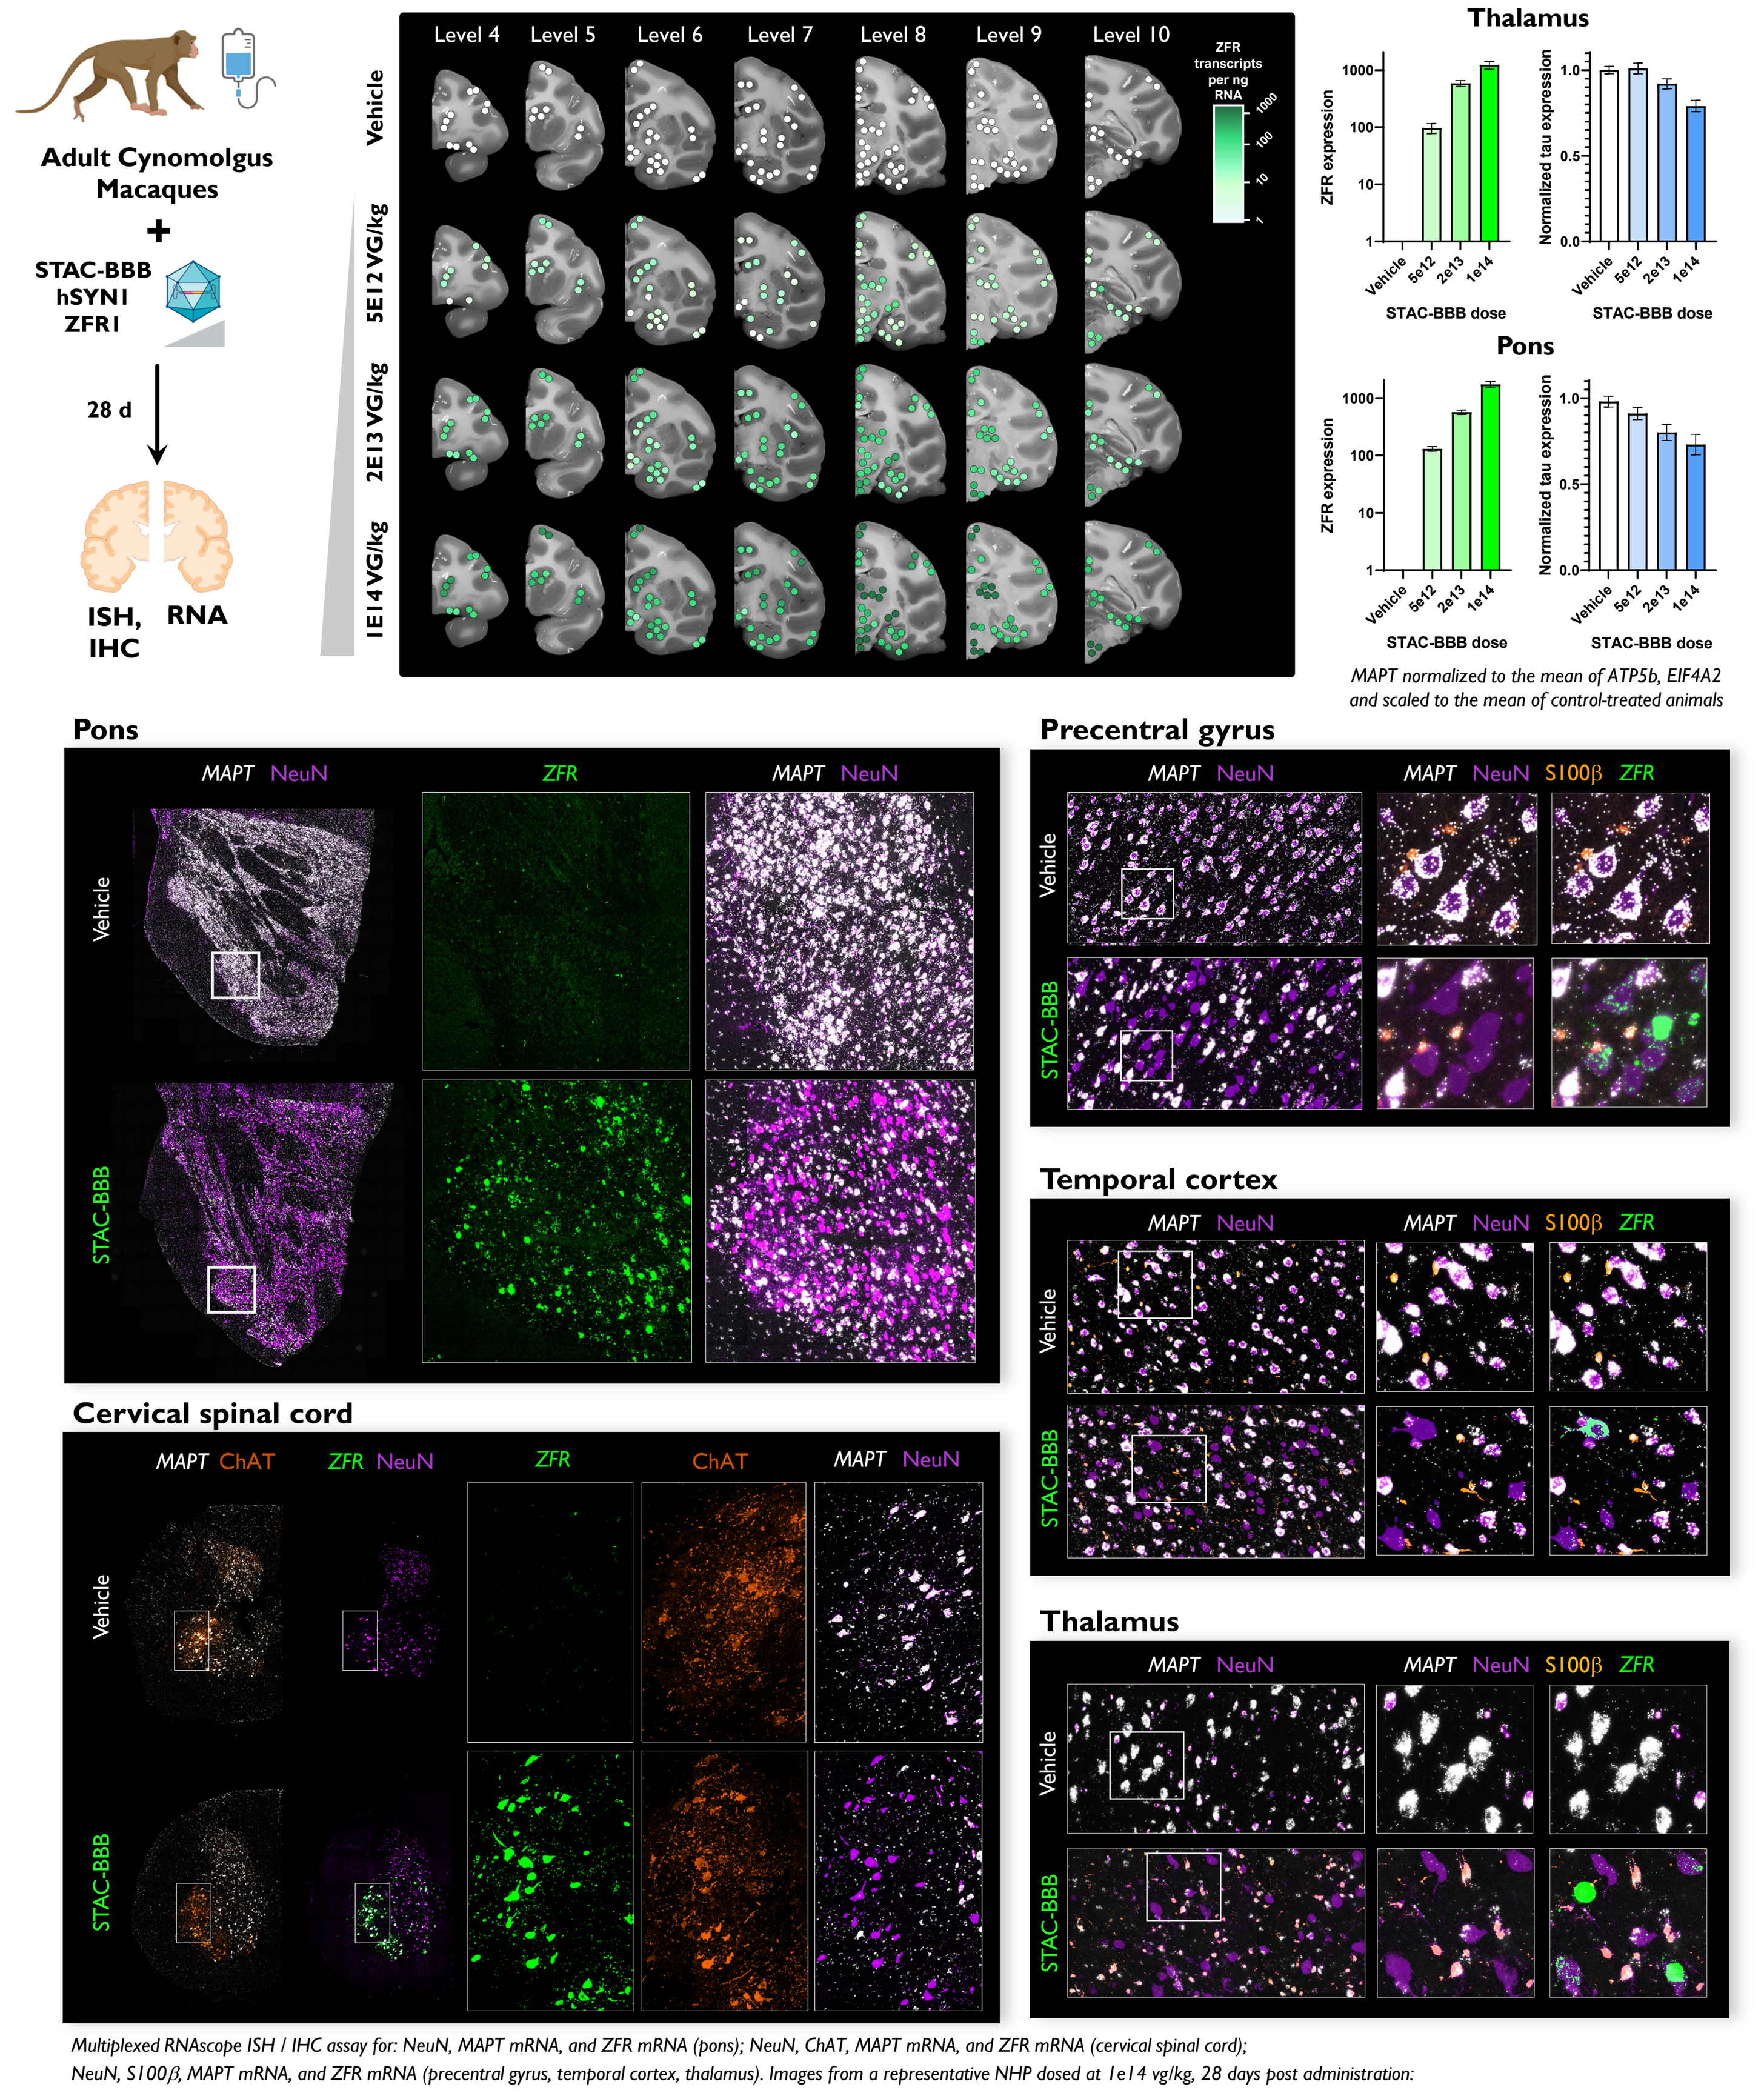
Adult Cynomolgus Macaques
STAC-BBB hSYN1 ZFR1
28 d
ISH, IHC
RNA
Level 4
Level 5
Level 6
Level 7
Level 8
Level 9
Level 10
ZFR transcripts per ng RNA
Vehicle
5E12 VG/kg
2E13 VG/kg
1E14 VG/kg
Thalamus
Pons
ZFR expression
Normalized tau expression
STAC-BBB dose
MAPT normalized to the mean of ATP5b, EIF4A2 and scaled to the mean of control-treated animals
Pons
MAPT NeuN
ZFR
STAC-BBB
Precentral gyrus
MAPT NeuN S100β ZFR
Cervical spinal cord
MAPT ChAT
ZFR NeuN
ChAT
Temporal cortex
Thalamus
Multiplexed RNAscope ISH / IHC assay for: NeuN, MAPT mRNA, and ZFR mRNA (pons); NeuN, ChAT, MAPT mRNA, and ZFR mRNA (cervical spinal cord); NeuN, S100β, MAPT mRNA, and ZFR mRNA (precentral gyrus, temporal cortex, thalamus). Images from a representative NHP dosed at 1e14 vg/kg, 28 days post administration.

Figure 7. Bulk and single-cell ZFR expression and *MAPT* repression across the NHP CNS

ZFR1 was delivered intravenously to adult NHPs using a novel BBB-penetrant capsid, STAC-BBB. Bulk RT-qPCR analysis of 220 punches per NHP showed dose-dependent ZFR expression throughout the entire brain at all dose levels. Dose-dependent *MAPT* repression was observed in many regions including the pons and thalamus. *MAPT* expression from non-neuronal cells contributes to this bulk signal. Single-cell analysis revealed widespread ZFR expression and neuron-specific *MAPT* repression throughout the brain and all spinal cord levels. This includes regions critical for AD and PSP.

Acknowledgments and disclosures

We would like to thank our previous collaborators, including Biogen and the laboratory of Dr. Brad Hyman. We also thank all current and former Sangamo colleagues that have contributed to the tau program. This work was funded by Sangamo Therapeutics.

Select images made with Biorender.com

Presented at ASGCT 2024